Hyper-sialylated IgG M254, an Innovative Therapeutic Candidate, Evaluated in Healthy Volunteers and Patients With Immune Thrombocytopenia Purpura: Safety, Tolerability, Pharmacokinetics, and Pharmacodynamics

1090

Santiago Arroyo, MD, PhD[1]; Renger G. Tiessen, MD, PhD[2]; William S. Denney, PhD[3]; Jim Jin, MS[1]; Mattheus Paulus (Thijs) van Iersel, MD[2]; Heidi Zeitz, MEd, CCRA[1]; Anthony M. Manning, PhD[1]; Martin R. Schipperus, MD, PhD[4]; James B. Bussel, MD[5]

[1]Momenta Pharmaceuticals, Inc, Cambridge, MA; [2]PRA Health Sciences, Groningen, Netherlands; [3]Human Predictions, LLC, Cambridge, MA; [4]Haga Teaching Hospital, The Hague, Netherlands; [5]Division of Pediatric Hematology/Oncology, Department of Pediatrics, New York Presbyterian Hospital Weill Cornell Medical Center, New York, NY

BACKGROUND

Intravenous immunoglobulin (IVIg), a therapeutic blood product prepared from pooled plasma of 3000–60,000 healthy donors, is the treatment of choice for several autoimmune disorders and immunodeficiency syndromes.

Although clinically useful, IVIg has several limitations including infusion-related adverse events, prolonged infusion times (1 day to over several days), large volumes required with a risk of volume overload, high cost, and supply shortages. IVIg alternatives could leverage the broad biological activities of IVIg and provide more consistent and potent anti-inflammatory activity with increased ease of administration.

M254, a novel hyper-sialylated immunoglobulin G (IgG) investigational product derived from commercially available IVIg, is hypothesized to have greater potency than IVIg (**Figure 1**). M254 was designed based on scientific evidence demonstrating the relevance of glycosylation on immunoglobulin function, especially the anti-inflammatory activity of IVIg, which has been shown to be dependent on Fc-sialylation. Preclinical data suggest that M254 has up to an ~10-fold enhancement of activity in 4 independent in vivo model systems: collagen antibody-induced arthritis and pemphigoid skin blistering models; K/BxN-sera–induced arthritis model; and an immune thrombocytopenia (ITP) mouse model.[1] These preclinical findings support the clinical evaluation of M254 in healthy volunteers and in patients with ITP.

Figure 1. Rationale for M254, a Hyper-sialylation of IVIg (hsIVIg)

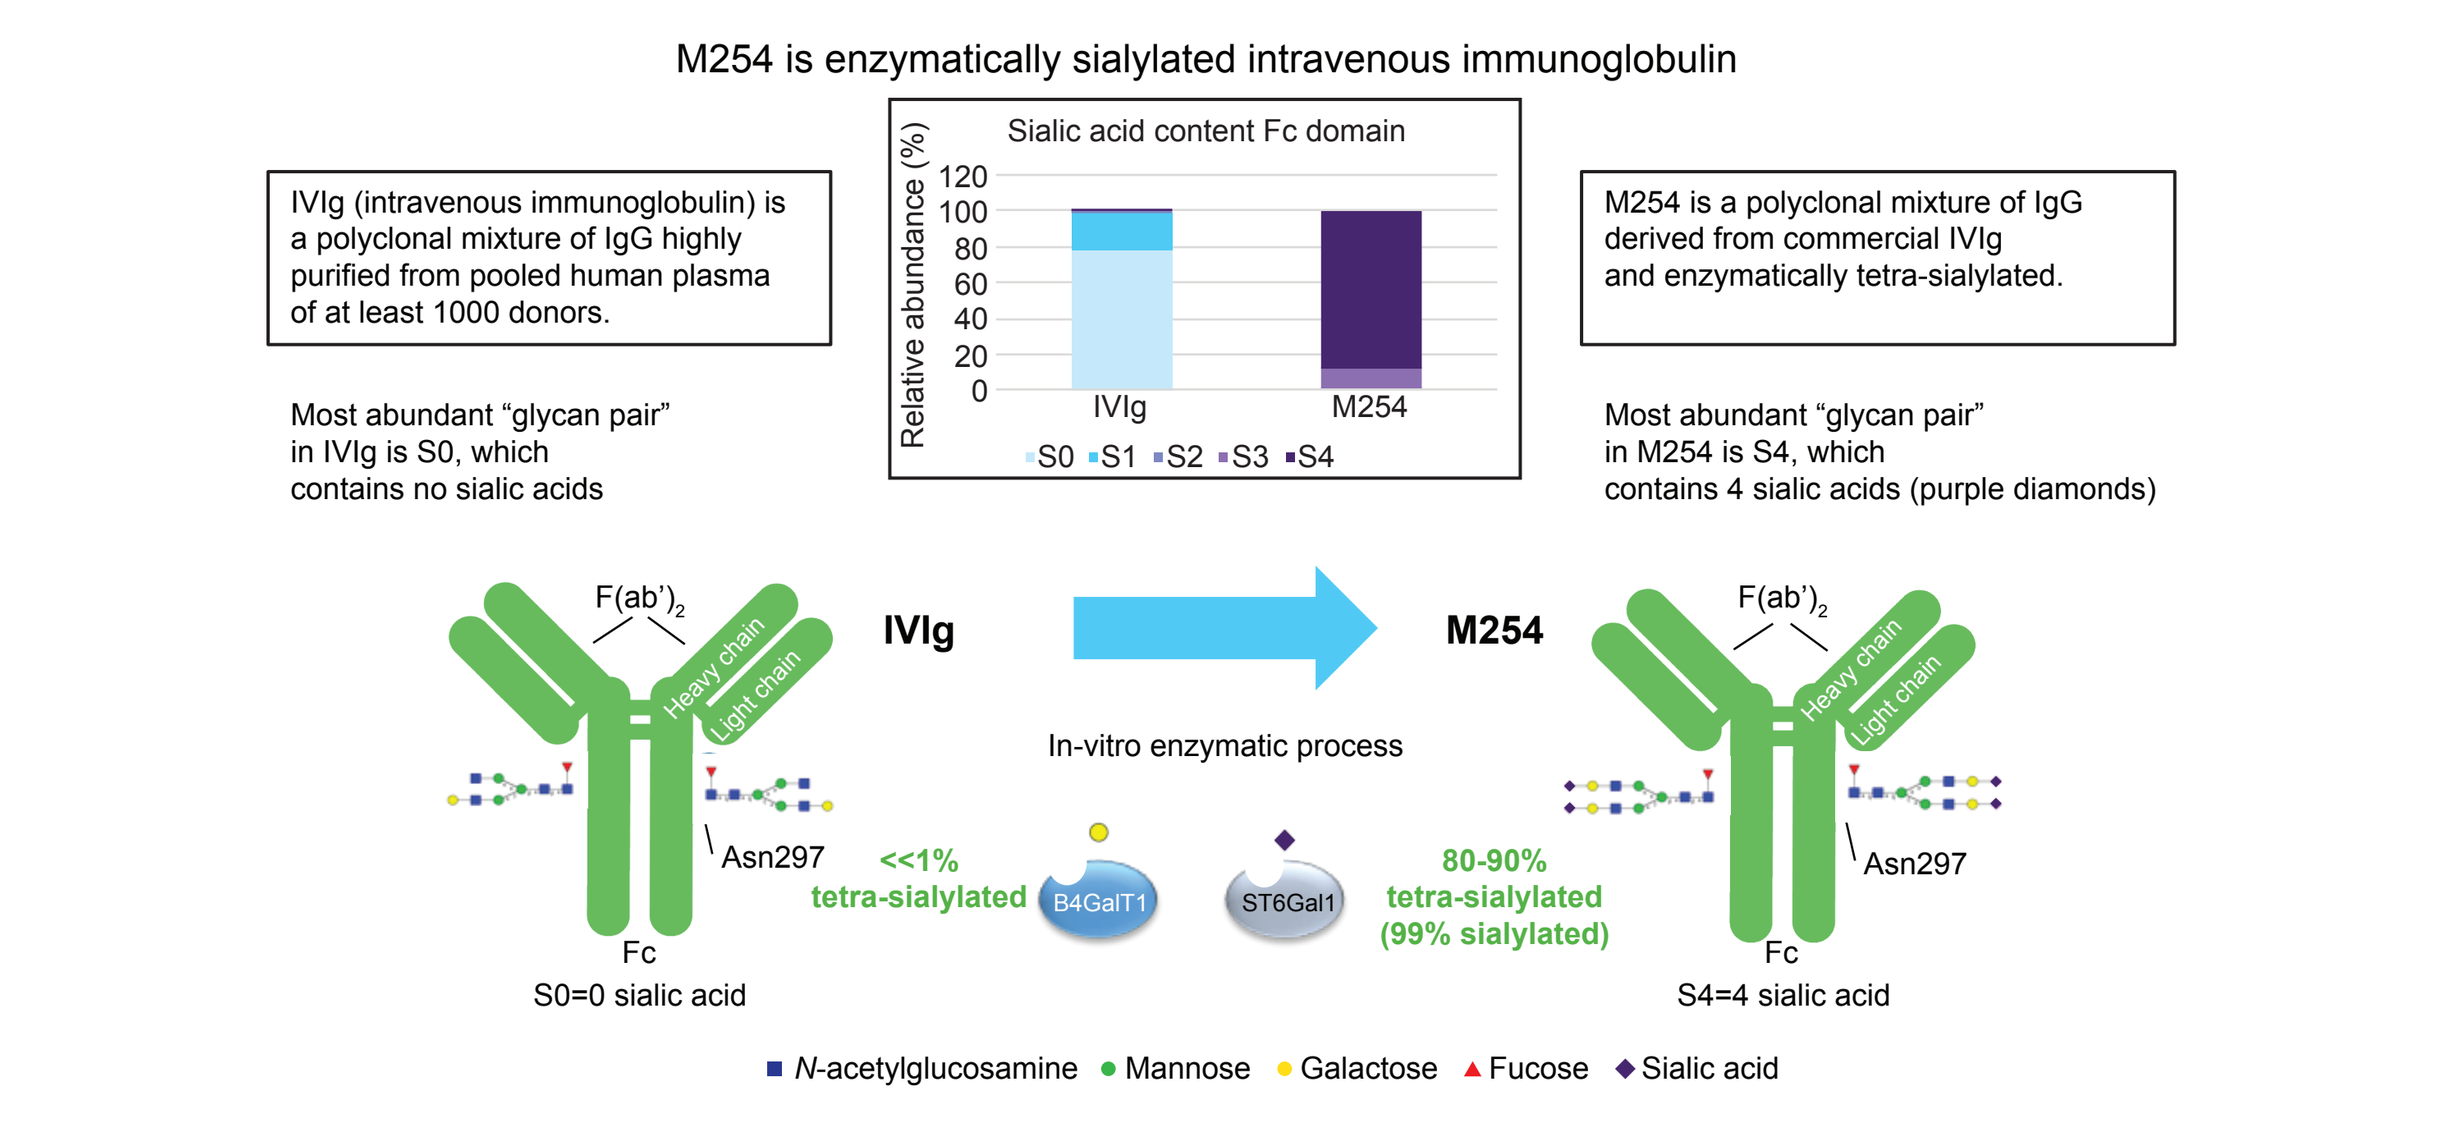
M254 is enzymatically sialylated intravenous immunoglobulin
IVIg (intravenous immunoglobulin) is a polyclonal mixture of IgG highly purified from pooled human plasma of at least 1000 donors.
Sialic acid content Fc domain
Relative abundance (%)
120
100
80
60
40
20
0
IVIg
M254
S0 S1 S2 S3 S4
M254 is a polyclonal mixture of IgG derived from commercial IVIg and enzymatically tetra-sialylated.
Most abundant "glycan pair" in IVIg is S0, which contains no sialic acids
Most abundant "glycan pair" in M254 is S4, which contains 4 sialic acids (purple diamonds)
F(ab')2
Heavy chain
Light chain
IVIg
M254
In-vitro enzymatic process
Asn297
<<1% tetra-sialylated
B4GalT1
ST6Gal1
80-90% tetra-sialylated (99% sialylated)
Fc
S0=0 sialic acid
S4=4 sialic acid
N-acetylglucosamine
Mannose
Galactose
Fucose
Sialic acid

IVIg, intravenous immunoglobulin

STUDY DESIGN AND METHODS

This first-in-human phase 1/2 clinical study is designed to evaluate the safety, tolerability, pharmacokinetics (PK), and pharmacodynamics (PD) of M254 compared with IVIg, and has 4 parts (NCT03866577). Part A enrolled 25 healthy volunteers; Parts B (ongoing), C, and D of the study will enroll patients with ITP. The study design is shown in **Figure 2**.

Figure 2. Overall Study Design

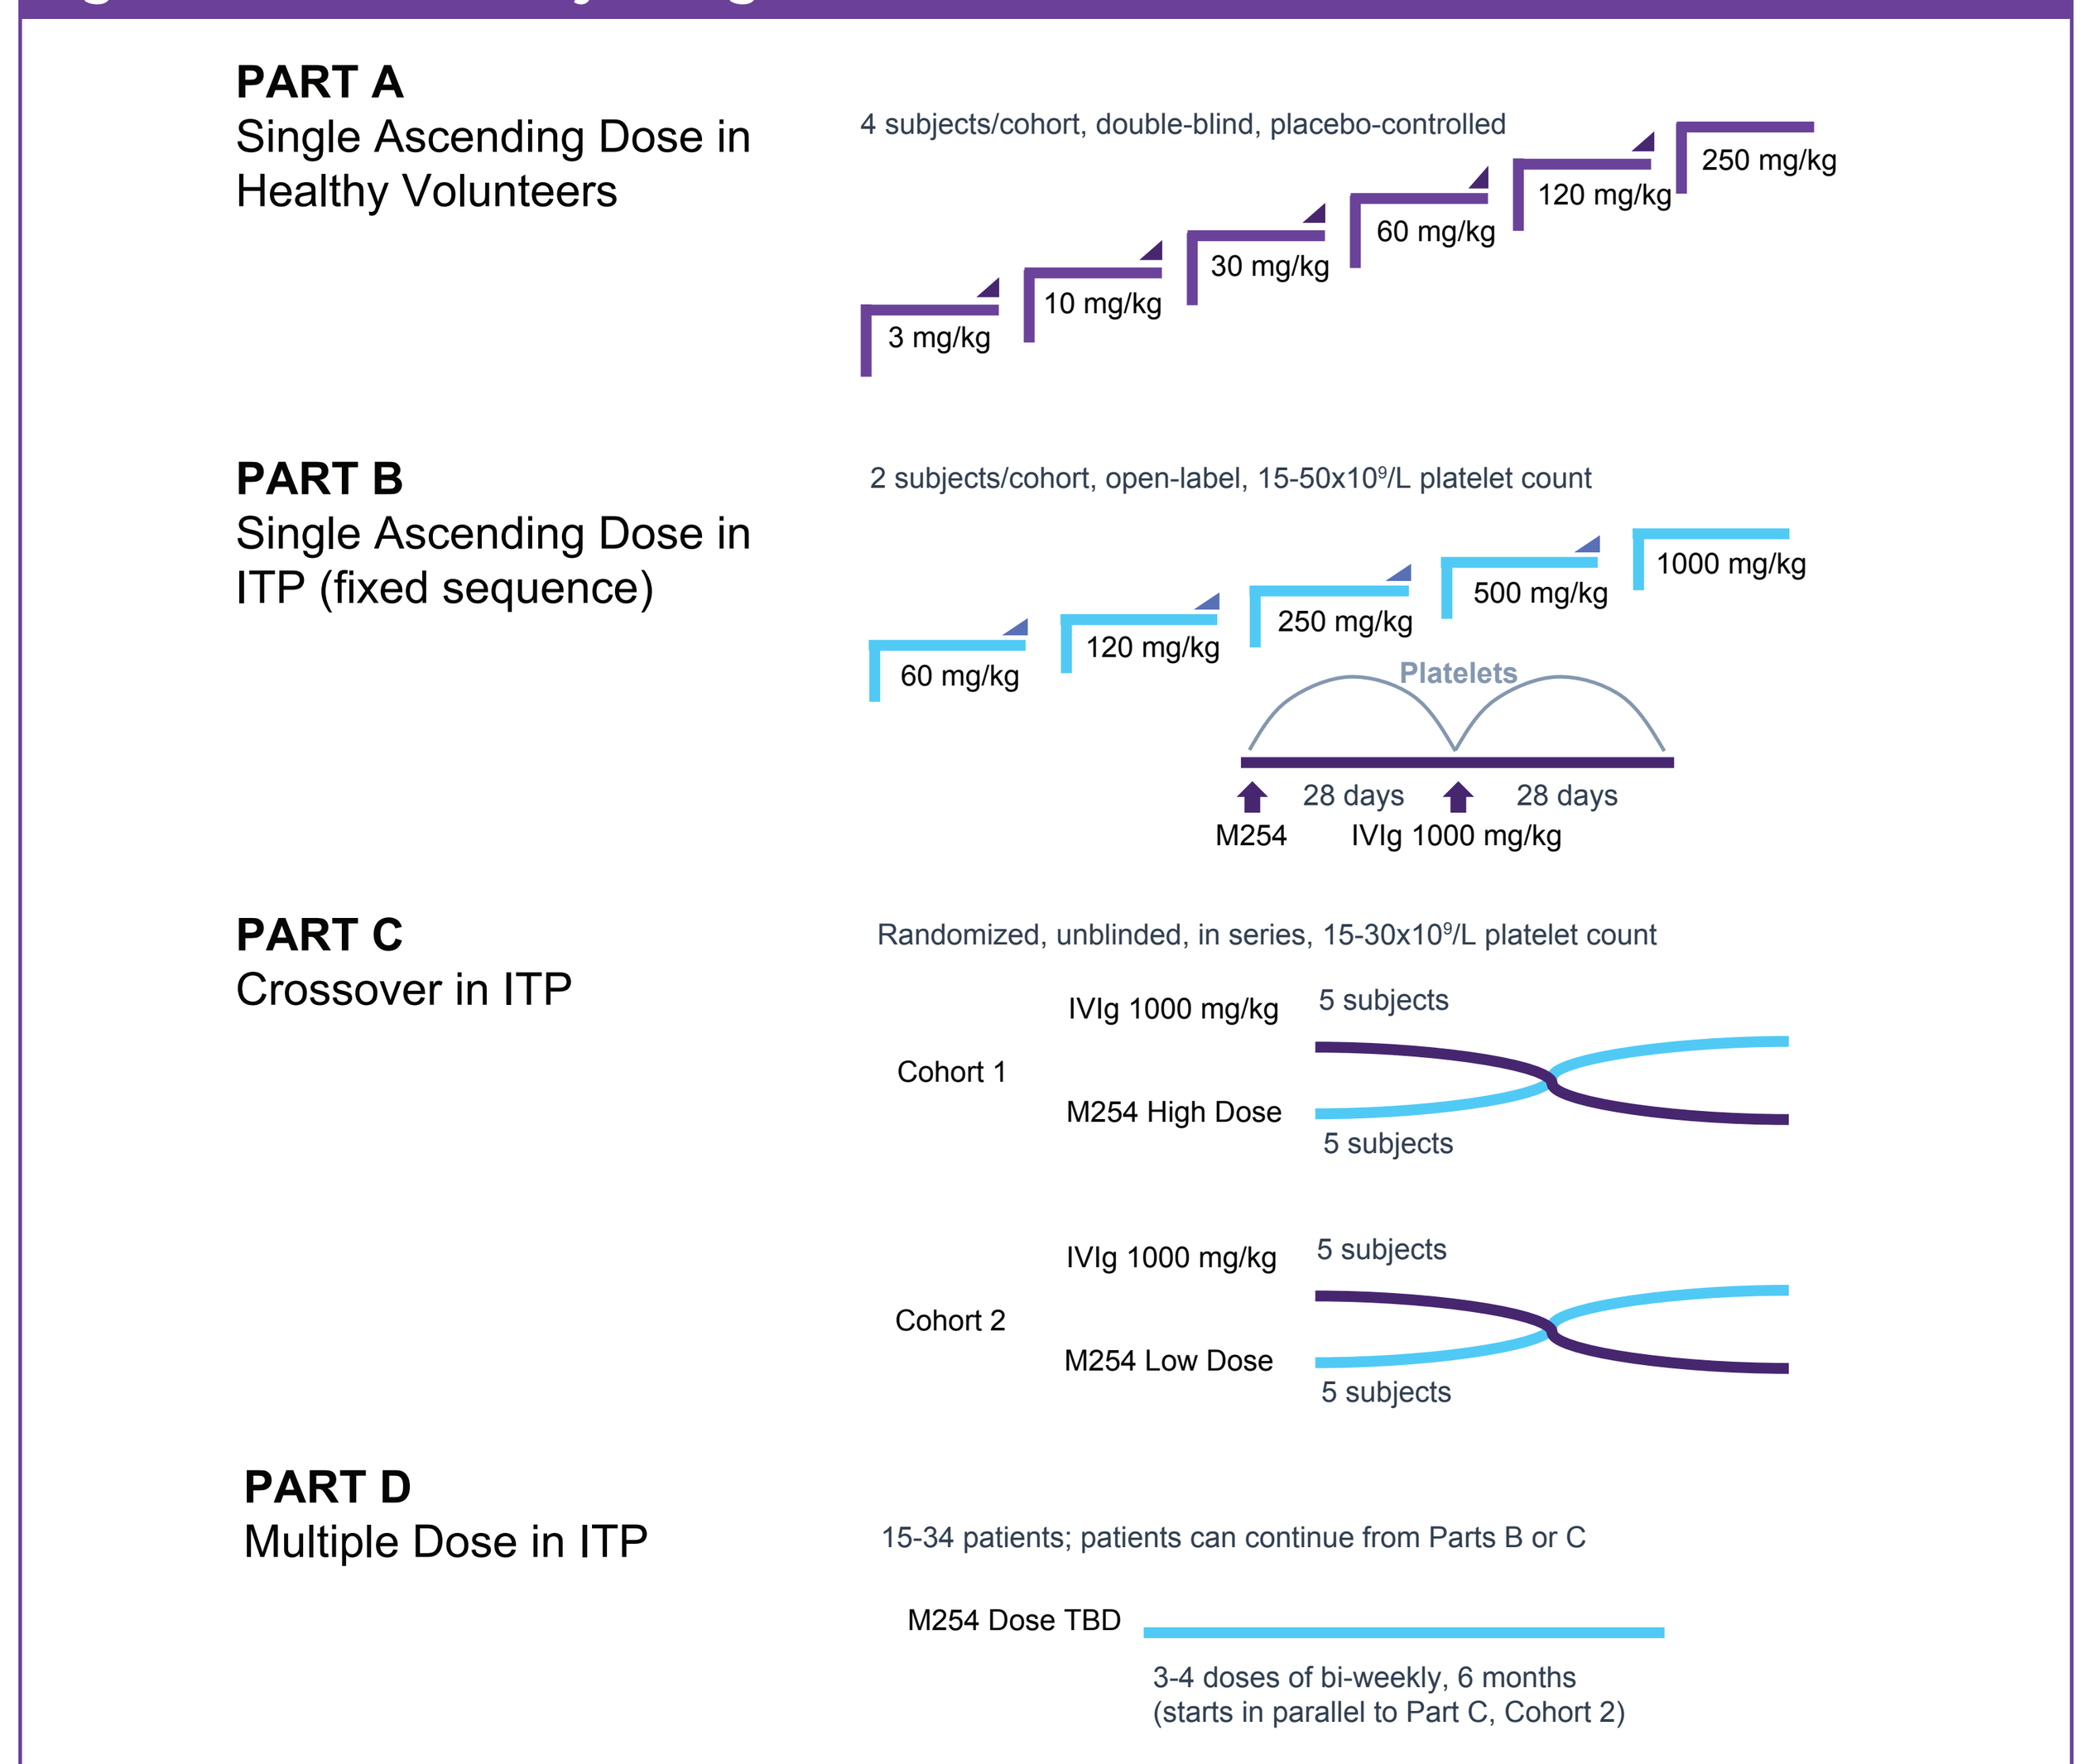
PART A
Single Ascending Dose in Healthy Volunteers
4 subjects/cohort, double-blind, placebo-controlled
3 mg/kg
10 mg/kg
30 mg/kg
60 mg/kg
120 mg/kg
250 mg/kg
PART B
Single Ascending Dose in ITP (fixed sequence)
2 subjects/cohort, open-label, 15-50x10⁹/L platelet count
60 mg/kg
120 mg/kg
250 mg/kg
500 mg/kg
1000 mg/kg
Platelets
28 days
28 days
M254
IVIg 1000 mg/kg
PART C
Crossover in ITP
Randomized, unblinded, in series, 15-30x10⁹/L platelet count
IVIg 1000 mg/kg
5 subjects
Cohort 1
M254 High Dose
5 subjects
IVIg 1000 mg/kg
5 subjects
Cohort 2
M254 Low Dose
5 subjects
PART D
Multiple Dose in ITP
15-34 patients; patients can continue from Parts B or C
M254 Dose TBD
3-4 doses of bi-weekly, 6 months
(starts in parallel to Part C, Cohort 2)

ITP, immune thrombocytopenia; IVIg, intravenous immunoglobulin

STUDY OBJECTIVE AND ENDPOINTS

Part A assessed safety, tolerability, and PK[2] of M254 after administration of a single ascending dose in healthy volunteers (n=25).

Part B assesses safety, tolerability, and PK of M254 after administration of a single ascending dose followed by 1000 mg/kg IVIg administration in patients with ITP (n=10), with an exploratory evaluation of platelet response with M254 compared with IVIg. This exploratory platelet-response evaluation allows a model-based, informed dose selection for Part C.

Part C will be a 2-arm crossover design comparing the platelet response of M254 with IVIg in patients with ITP (n=20). Part C will have 2 cohorts: high dose M254 in Cohort 1 and low dose M254 in Cohort 2. In combination with Part B, Part C will allow a model-based estimate of the M254 dose that is equivalent to IVIg. Using the totality of the data as well as dose ranging will enable high confidence in the M254 dose while requiring a minimum number of patients with ITP.

Part D will be a multiple dose study of M254 for the evaluation of safety, tolerability, PK, and PD platelet response in patients with ITP.

ELIGIBILITY CRITERIA FOR STUDY PARTICIPATION

Part A: Healthy Volunteers

Key Inclusion Criteria	Key Exclusion Criteria
• Men or women aged 18 to 55 years • Good health as indicated by medical history, physical examination, vital signs, clinical laboratory tests, 12-lead ECG • Capable of giving signed informed consent	• History of any drug allergy, hypersensitivity, or intolerance to any drug product • History or presence of clinically significant medical conditions • Any illness within 5 days

Parts B, C, and D: Adult Patients With Immune Thrombocytopenic Purpura (ITP)

Key Inclusion Criteria	Key Exclusion Criteria
• Men or women aged 18 to 64 years • Diagnosed with ITP (with or without splenectomy) for ≥3 months • Platelet count between 15 and 50 x 10^9/L • Receiving stable (≥4 weeks) maintenance immunosuppressive therapy	• Newly diagnosed ITP patients who are naïve to anti-ITP treatments • History of arterial or venous thrombosis • Having ≥2 risk factors for thrombosis or clotting disorders • Known history of nonresponse to IVIg • Patients with severe or clinically significant bleeding (eg, grade 3 for skin, ≥ grade 2 for mucosal domains, or ≥ grade 1 for organ domain using the ITP-BAT) • Patients taking thrombopoietin receptor agonist AND have albumin ≤35 g/L • Receiving rituximab within 3 months of screening

ECG, electrocardiogram; ITP-BAT, immune thrombocytopenia bleeding assessment tool

TRIAL PROGRESS AND UPDATES

Part A (healthy volunteers) has been completed. Results are presented below. Based on the evidence for tolerability in Part A, the study has progressed to Part B (patients with ITP).

Results From Part A: Healthy Volunteers

Subject Disposition

A total of 90 subjects were screened, and 25 of those subjects were included in Part A of the study. No subjects were withdrawn and none of the subjects dropped out during the course of the study. All subjects completed the study as per protocol.

- Group 1: a single IV infusion of 3 mg/kg M254 (n=3) or matching placebo (n=2)
- Group 2: a single IV infusion of 10 mg/kg M254 (n=3) or matching placebo (n=1)
- Group 3: a single IV infusion of 30 mg/kg M254 (n=3) or matching placebo (n=1)
- Group 4: a single IV infusion of 60 mg/kg M254 (n=3) or matching placebo (n=1)
- Group 5: a single IV infusion of 120 mg/kg M254 (n=3) or matching placebo (n=1)
- Group 6: a single IV infusion of 250 mg/kg M254 (n=3) or matching placebo (n=1)

Demographic Characteristics

A total of 13 female (52%) and 12 male (48%) subjects between 20 and 51 years of age and with a body mass index between 20.3 and 28.8 kg/m[2] participated in Part A of the study. Twenty subjects (80%) were white, 2 subjects (8%) were Asian, 1 subject (4%) was black or African American, 1 subject (4%) was American Indian or Alaska Native, and 1 subject (4%) was of multiple race. Two (8%) of the 25 subjects were of Hispanic or Latino ethnicity.

Pharmacokinetics

Figure 3. Median M254 Plasma Concentrations versus Time Profile

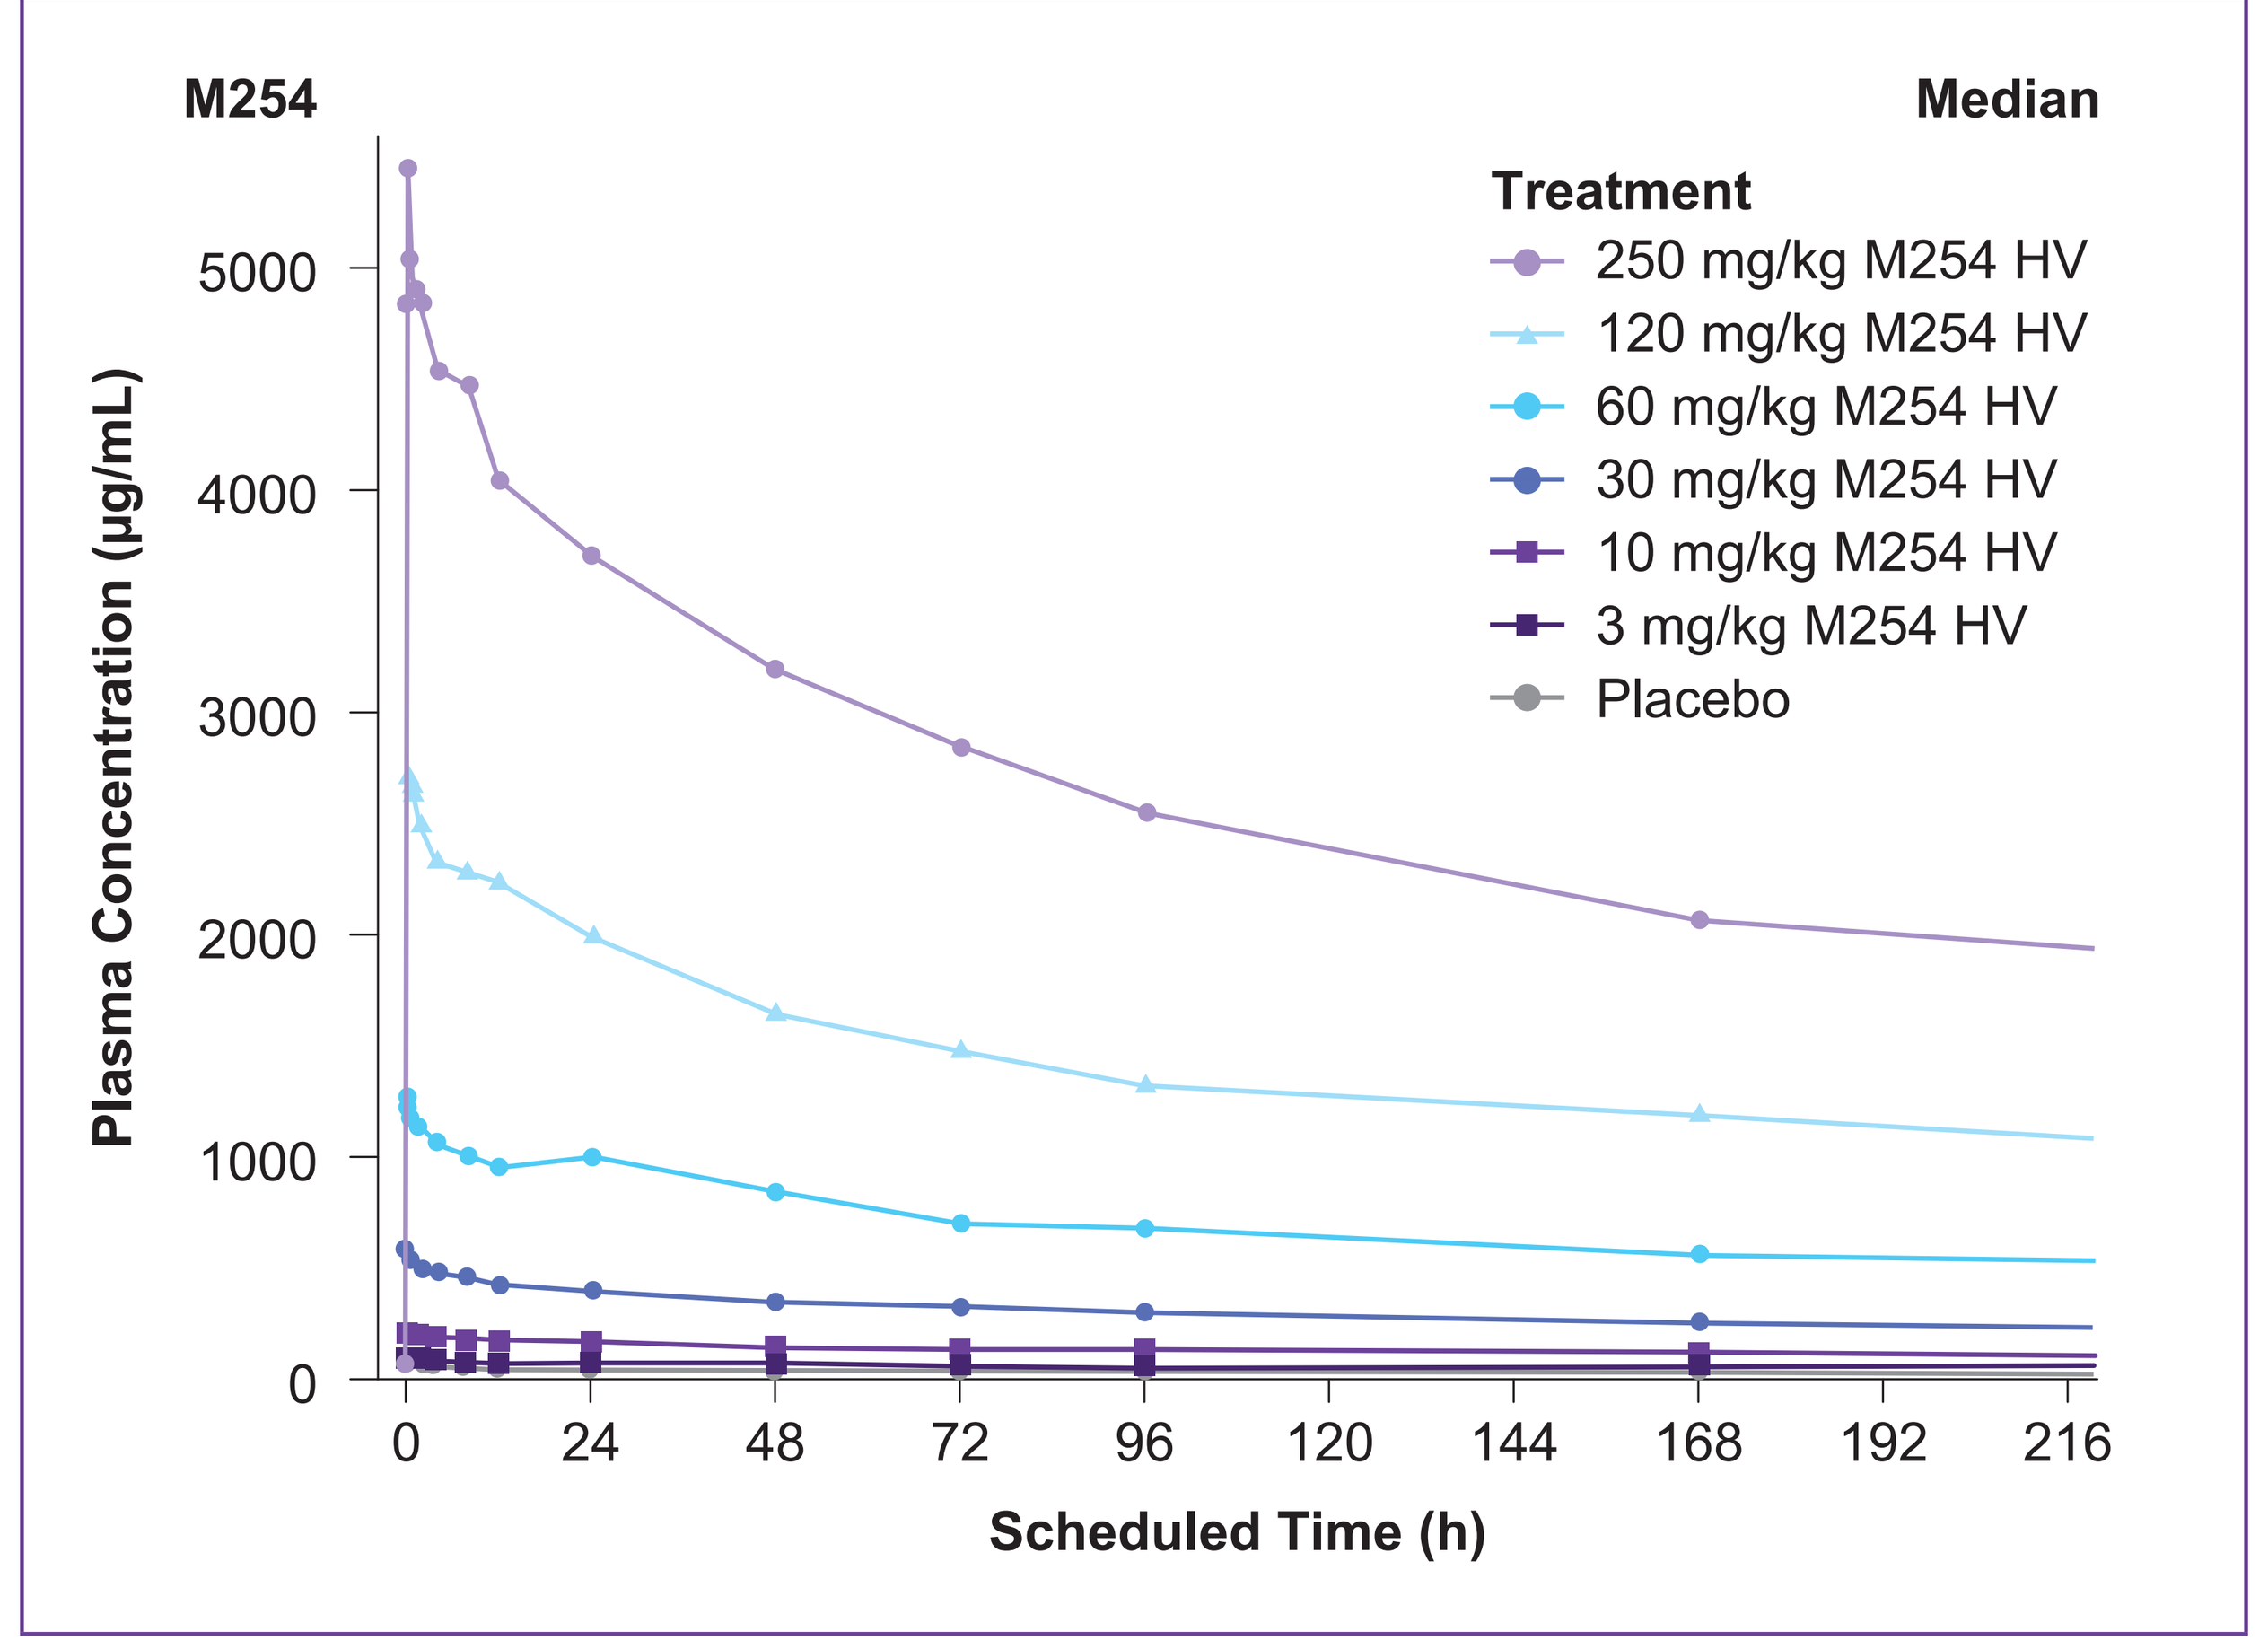
M254
Median
Treatment
250 mg/kg M254 HV
120 mg/kg M254 HV
60 mg/kg M254 HV
30 mg/kg M254 HV
10 mg/kg M254 HV
3 mg/kg M254 HV
Placebo
Plasma Concentration (µg/mL)
0
1000
2000
3000
4000
5000
0 24 48 72 96 120 144 168 192 216
Scheduled Time (h)

- The M254 plasma concentration linearly increased with M254 dose (**Figure 3**).
- Measurable M254 concentrations were still observed on Day 22 over the dose range from 3 mg/kg to 60 mg/kg and up to Day 29±2 in the 120 mg/kg and 250 mg/kg dose groups.
- Maximum serum concentration (C_{max}) and area under the curve (AUC) are approximately dose-proportional, and half-life appears similar to the half-life reported for IgG after IVIg infusion.

Safety

- There were no serious or severe adverse events or deaths.
- The majority of the treatment-emergent adverse events were mild, transient, and resolved without intervention.
- One subject in the 250 mg/kg M254 group had a transient moderate infusion reaction and study drug administration was stopped prematurely, so this subject received 174 mg/kg only.
- Single IV doses of M254 ranging from 3 mg/kg to 250 mg/kg appeared to be safe and well tolerated in healthy male and female subjects in Part A.

Summary of Related Treatment-Emergent Adverse Events by Preferred Terms in Healthy Volunteers

		M254 Dose Groups						
Parameter	Placebo (N=7) n	3 mg/kg (N=3) n	10 mg/kg (N=3) n	30 mg/kg (N=3) n	60 mg/kg (N=3) n	120 mg/kg (N=3) n	250 mg/kg (N=3) n	All (N=18) n
Total	3	1	2	2			3	8
Abdominal pain			1					1
Diarrhea			1					1
Soft feces			1					1
Nausea	2						1	1
Vomiting							1	1
Chills	1						1	1
Fatigue	1							
Pyrexia							1	1
Back pain	1							
Dizziness	1							
Headache	1	1		1			3	5
Nasal congestion			1					1
Nasal dryness			1					1
Flushing				1				1

N/n = number of subjects with events

PART B, C, and D – PK-PD Assessment

Part B will estimate the initial dose- or exposure-response for platelet changes following M254 administration. Because of the small number of patients included in Part B, Part B will allow only an initial estimation of dose- or exposure-response. Patients in Part B will also receive a fixed dose of 1 g/kg IVIg for comparison because of the inherent variability in platelet response to IVIg.

From the initial M254 to IVIg potency ratio estimates in Part B, Part C will improve the precision of the estimate of the ratio of M254 to IVIg dose required to achieve the desired platelet response. The improved precision from Part C is anticipated to be above and below the M254 dose equivalent to 1 g/kg IVIg.

Part D will further improve the understanding of M254 platelet-response efficacy, and will inform multi-dose safety and efficacy of M254.

Illustrations of potential platelet-response precision compared with 1 g/kg IVIg following Parts B and C are shown below (**Figure 4**). These figures are illustrations only.

Figure 4. Illustrations Comparing the Potential Platelet Response Between IVIg and M254

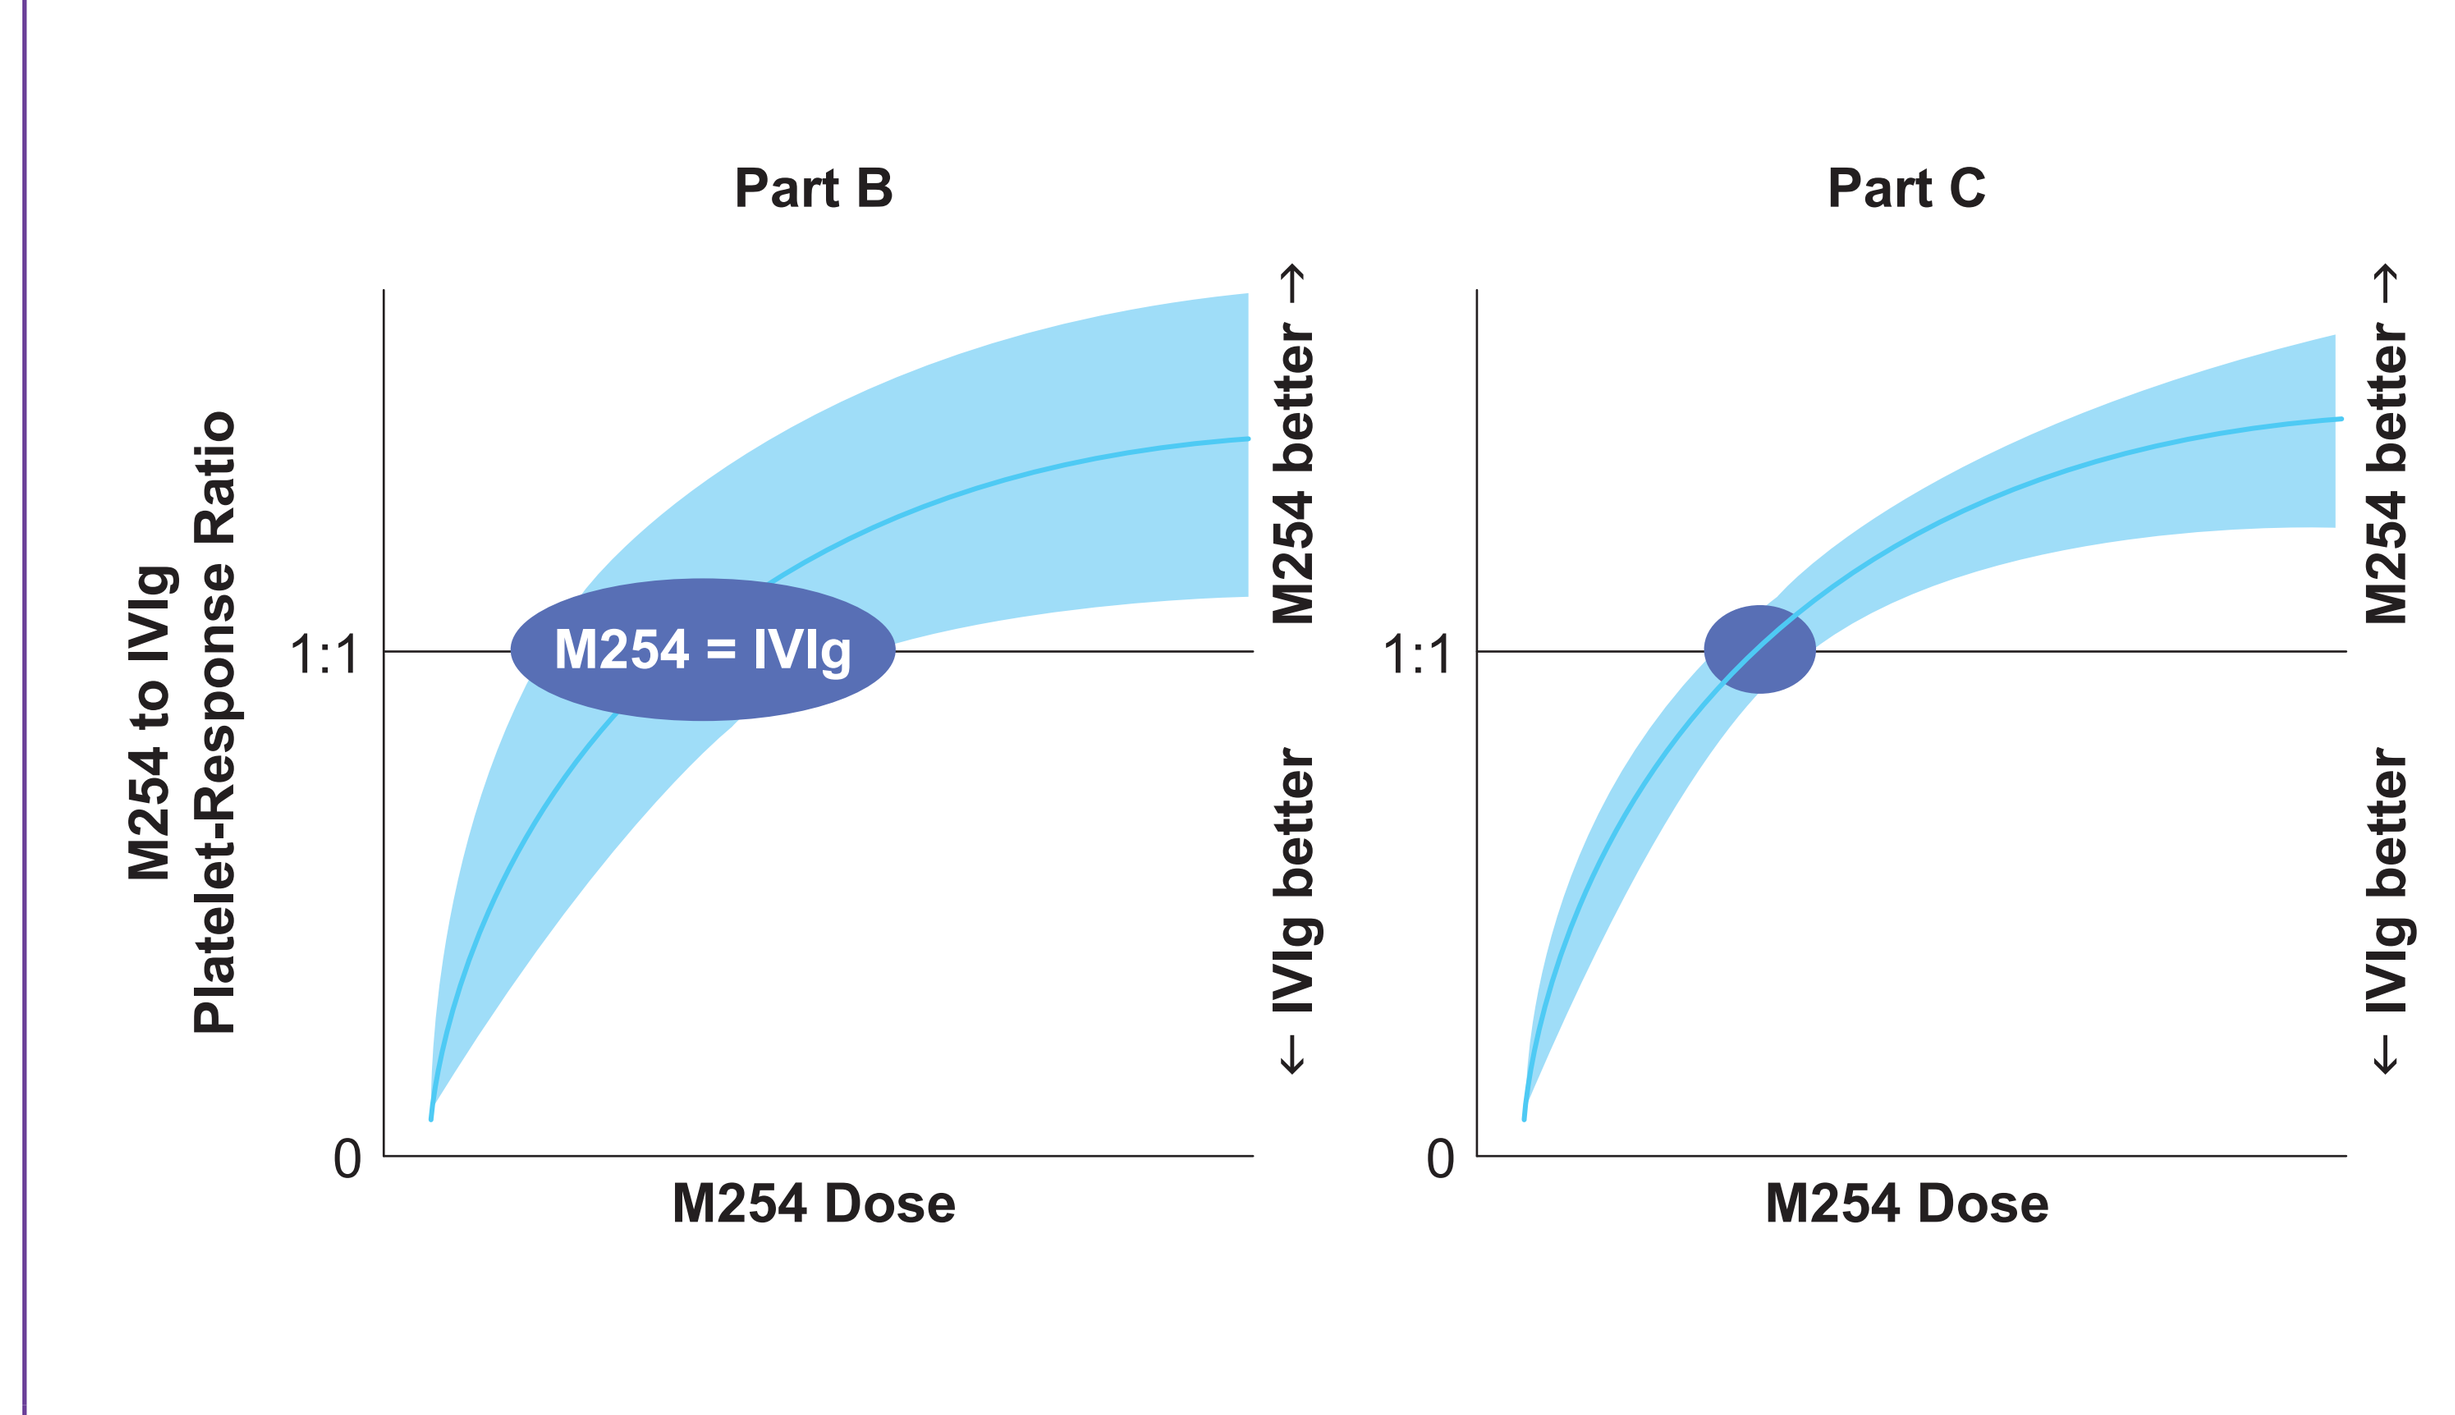
Part B
Part C
M254 to IVIg Platelet-Response Ratio
1:1
0
M254 = IVIg
M254 better →
← IVIg better
M254 Dose

IVIg, intravenous immunoglobulin

SUMMARY

M254 is a novel hyper-sialylated IgG therapeutic candidate derived from commercially available IVIg, with potentially greater potency when compared with IVIg on an mg/kg basis. The present 4-part first-in-human study is being conducted first in healthy volunteers and then in patients with ITP to evaluate the safety, tolerability, PK, and PD and platelet response to M254 administered as a single followed by multiple IV doses for both M254 itself and in comparison with IVIg.

Part A assessed the safety, tolerability, and PK of M254 after administration of single ascending doses of M254 in healthy volunteers. Overall, single IV doses of M254 ranging from 3 mg/kg to 250 mg/kg appeared to be safe and well tolerated by the majority of the male and female healthy volunteers. Most of the adverse events were mild, entirely different single events, and there was no increased occurrence of any specific adverse event with increasing dose. M254 serum concentrations increased with dose in a dose-linear manner. Upon single dosing with M254, the total IgG serum concentrations increased in the 250 mg/kg dose group and slightly increased in the 120 mg/kg dose group as expected, as M254 consists of a mixture of IgG subtypes.

The study is currently enrolling patients with ITP in Part B.

REFERENCES

1. Washburn N, Schwab I, Ortiz D, et al. Controlled tetra-Fc sialylation of IVIg results in a drug candidate with consistent enhanced anti-inflammatory activity. *PNAS*. 2015;112:E1297-E1306.
2. Teekamp N, Washburn N, Bronsema K, et al. LC-MS/MS quantification of M254, a hyper-sialylated endogenous IgG biotherapeutic: analytical pitfalls and solutions. Presented at the European Bioanalysis Forum; 20-22 November 2019; Barcelona, Spain.

ACKNOWLEDGMENTS

This study is being sponsored by Momenta Pharmaceuticals.

Presented at the 2019 American Society of Hematology Annual Meeting; Orlando, Florida, United States; December 7, 2019